Phase 1/2, first-in-human study of AVZO-103, a bispecific Nectin4/Trop2 antibody-drug conjugate (BsADC), as monotherapy and in combination therapy in patients with locally advanced or metastatic urothelial cancer (UC) or other solid tumors

Benjamin Garmezy, MD[1]; Andrae L. Vandross, MD[2]; Cesar A. Perez, MD[3]; Neal D. Shore, MD, FACS[4]; Daniel P. Petrylak, MD[5]; Mohammad Hirmand, MD[6]; Minal Mehta, MBBS[6]; Vadim S. Koshkin, MD[7]

[1]Sarah Cannon Research Institute, Nashville, TN, USA; [2]NEXT Oncology, Austin, TX, USA; [3]Florida Cancer Specialists and Research Institute, Orlando, FL, USA; [4]START Carolinas/Carolina Urologic Research Center, Myrtle Beach, SC, USA; [5]Yale Cancer Center, New Haven, CT, USA; [6]Clinical Development, Avenzo Therapeutics, Inc., San Diego, CA, USA; [7]Helen Diller Family Comprehensive Cancer Center, University of California San Francisco, San Francisco, CA, USA

FOR ADDITIONAL INFORMATION

TPS908

NCT07193511

BACKGROUND[1-11]

AVZO-103, a Novel BsADC That Selectively Targets Tumor Cells to Inhibit Growth

- Nectin4 and Trop2 are transmembrane proteins frequently overexpressed in UC and other solid tumors, but with limited co-expression in normal tissue[1-4]
- Single-target ADCs such as enfortumab vedotin (EV) have demonstrated clinical benefit[5,6]
 - However, current use is limited by resistance mechanisms and safety concerns[7-9]
- AVZO-103 is a potential best-in-class BsADC designed to selectively target tumor cells expressing Nectin4 and Trop2[10]

AVZO-103 Inhibits Tumor Growth in Preclinical Xenograft Models

- In preclinical xenograft studies including EV refractory models, AVZO-103 demonstrated robust tumor growth inhibition **(Figure 1)**[10]
- In tumor cells overexpressing both Nectin4 and Trop2, AVZO-103 demonstrated enhanced binding, internalization, and cytotoxicity
 - AVZO-103 has moderate affinity for both Nectin4 and Trop2, potentially reducing target-driven toxicities in normal tissues
- Combining AVZO-103 with an immunotherapy is supported by clinical validation of PD-1 inhibitor efficacy enhancement in Nectin4- and Trop2-expressing tumors, as well as preclinical data showing topoisomerase I-based ADCs enhance PD-1 inhibitor efficacy[11]

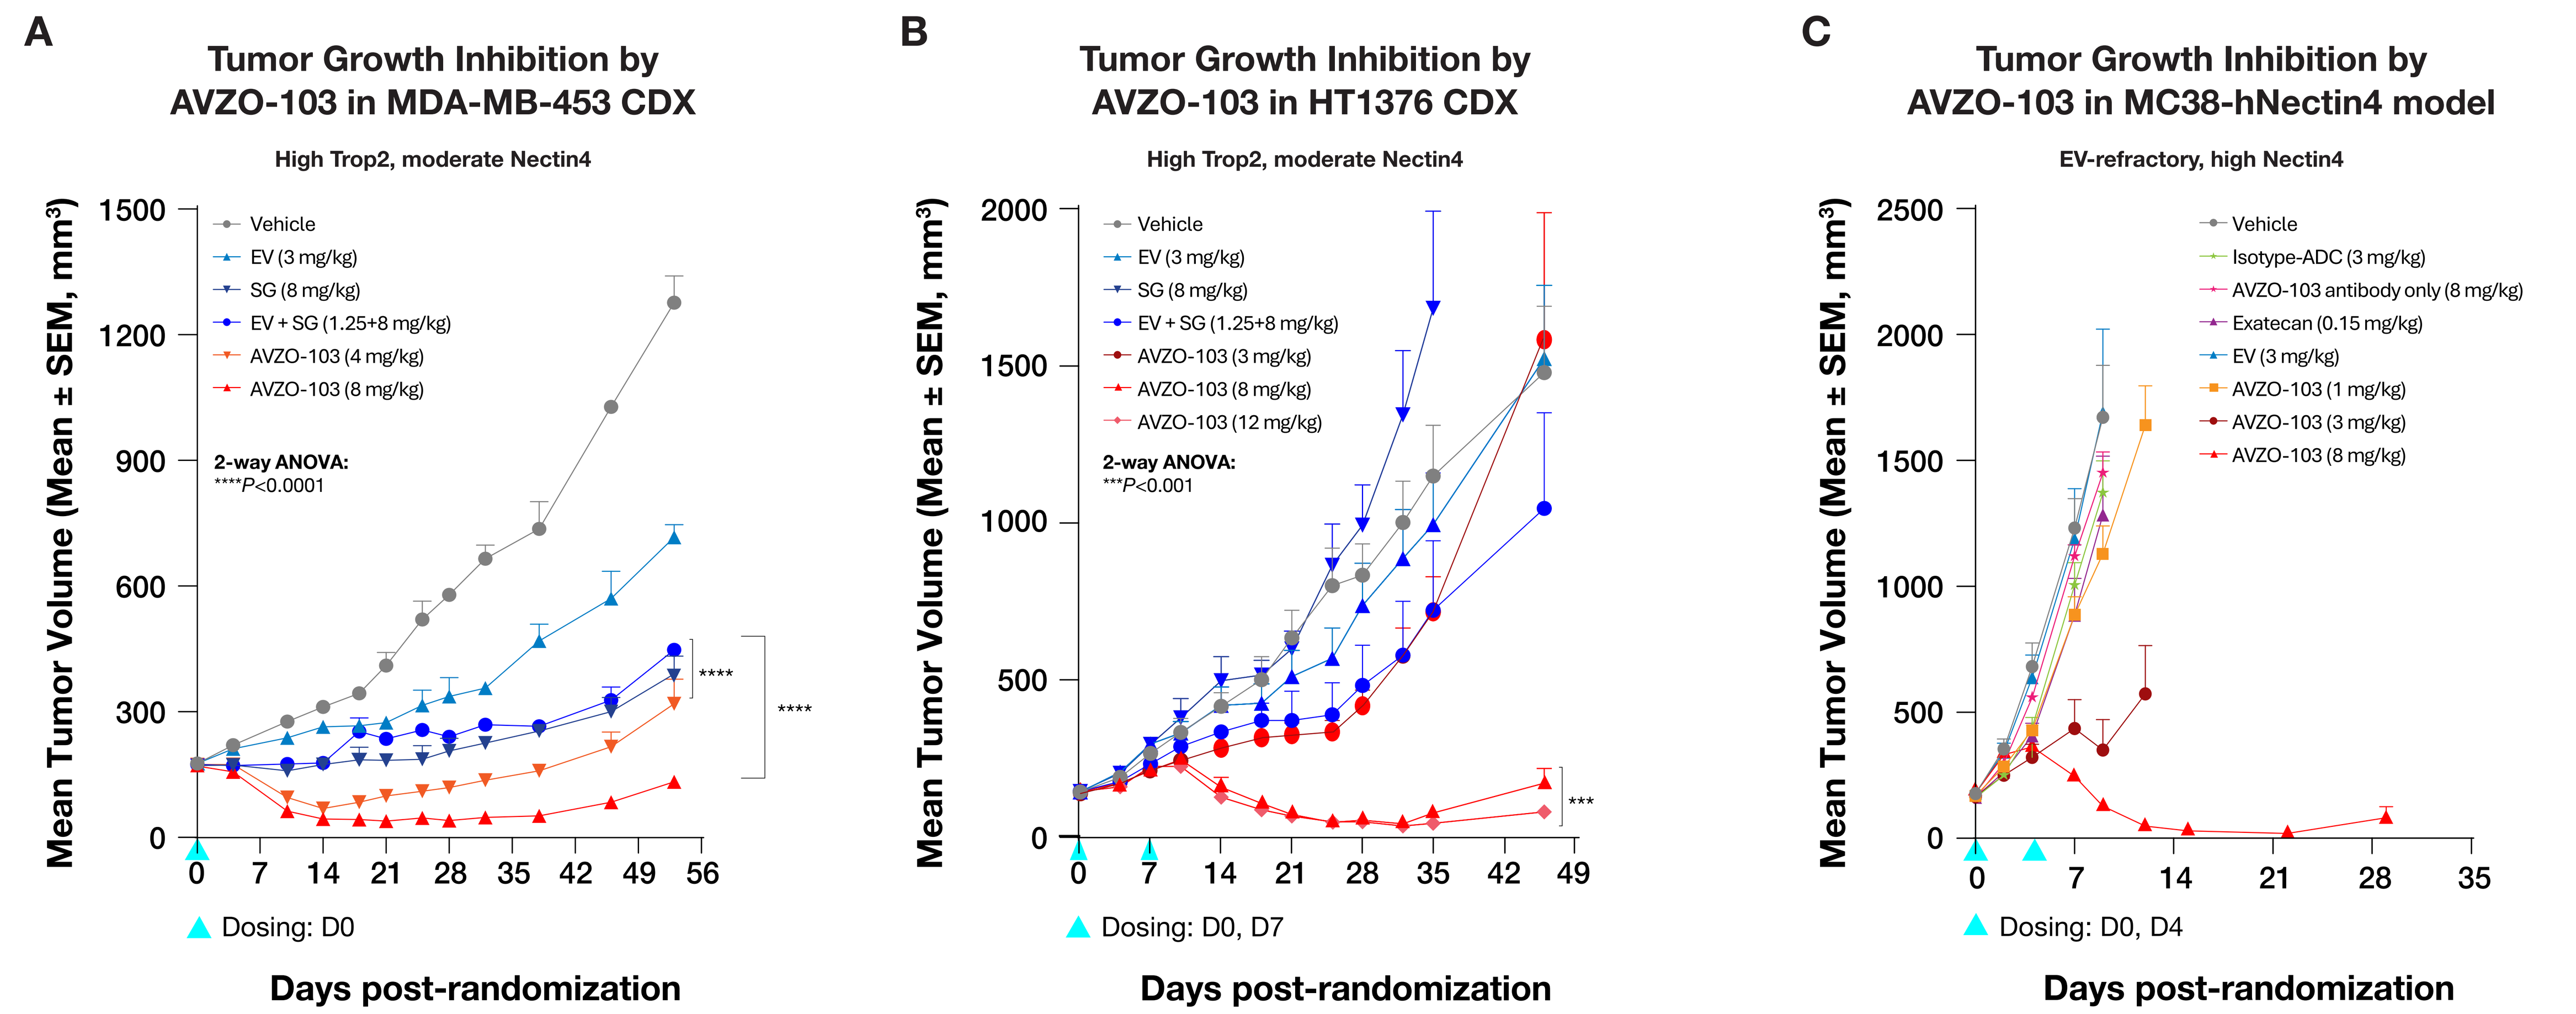
A
Tumor Growth Inhibition by AVZO-103 in MDA-MB-453 CDX
High Trop2, moderate Nectin4
Vehicle
EV (3 mg/kg)
SG (8 mg/kg)
EV + SG (1.25+8 mg/kg)
AVZO-103 (4 mg/kg)
AVZO-103 (8 mg/kg)
2-way ANOVA:
****P<0.0001
Dosing: D0
B
Tumor Growth Inhibition by AVZO-103 in HT1376 CDX
High Trop2, moderate Nectin4
Vehicle
EV (3 mg/kg)
SG (8 mg/kg)
EV + SG (1.25+8 mg/kg)
AVZO-103 (3 mg/kg)
AVZO-103 (8 mg/kg)
AVZO-103 (12 mg/kg)
2-way ANOVA:
***P<0.001
Dosing: D0, D7
C
Tumor Growth Inhibition by AVZO-103 in MC38-hNectin4 model
EV-refractory, high Nectin4
Vehicle
Isotype-ADC (3 mg/kg)
AVZO-103 antibody only (8 mg/kg)
Exatecan (0.15 mg/kg)
EV (3 mg/kg)
AVZO-103 (1 mg/kg)
AVZO-103 (3 mg/kg)
AVZO-103 (8 mg/kg)
Dosing: D0, D4
Mean Tumor Volume (Mean ± SEM, mm³)
Days post-randomization

ANOVA, analysis of variance; **CDX,** cell line-derived xenograft; **EV,** enfortumab vedotin; **SEM,** standard error of the mean; **SG,** sacituzumab govitecan.

Figure 1. AVZO-103 inhibited tumor growth in cell line-derived xenograft (CDX) models in a dose-dependent manner.[10]
Tumor growth inhibition in the human breast cancer MDA-MB-453 CDX model **(A)**. Tumor growth inhibition in the human bladder cancer HT1376 CDX model **(B)**. Tumor growth inhibition in the EV-resistant mouse colon cancer MC38-hNectin4 subcutaneous xenograft model **(C)**.

With its unique bispecific design, AVZO-103 allows for targeted delivery and controlled release of a potent topoisomerase I inhibitor, exatecan, to Nectin4- and Trop2-expressing cells. This may address tumor heterogeneity of target expression as well as overcome resistance to prior therapies.

AVZO-103-1001 STUDY DESIGN[10,12]

Study Objectives and Patient Population

- AVZO-103-1001 is a first-in-human Phase 1/2 study that aims to evaluate safety and tolerability when AVZO-103 is administered intravenously to patients with locally advanced or metastatic UC and other solid tumors known to express Nectin4
- Phase 1 of the study will assess AVZO-103 as monotherapy (Part A) and in combination with an immunotherapy (Part B) **(Figure 2)**
- Patients in Part A dose escalation will have locally advanced or metastatic solid tumors[a] known to express Nectin4 for whom standard therapies are no longer effective, appropriate, or declined by the patient
 - Backfill in Part A will enroll patients with locally advanced or metastatic UC

[a]Includes UC, cervical cancer, triple-negative breast cancer (TNBC), squamous cell carcinoma of the head and neck (SCCHN), non-squamous non-small cell lung cancer (NSCLC) with epidermal growth factor receptor (EGFR) mutations, and non-squamous NSCLC with no actionable genomic alterations.

Phase 1 Study Design

- Part A will enroll ~80 patients in an every-three-weeks escalating dosing schedule using a Bayesian Optimal Interval Design (BOIN) design to determine the maximum tolerated dose (MTD) and preliminary recommended Phase 2 dose (RP2D)
- Part B will enroll ~35 patients, with doses to be determined by safety review committee based on safety and pharmacokinetic (PK) data from Part A

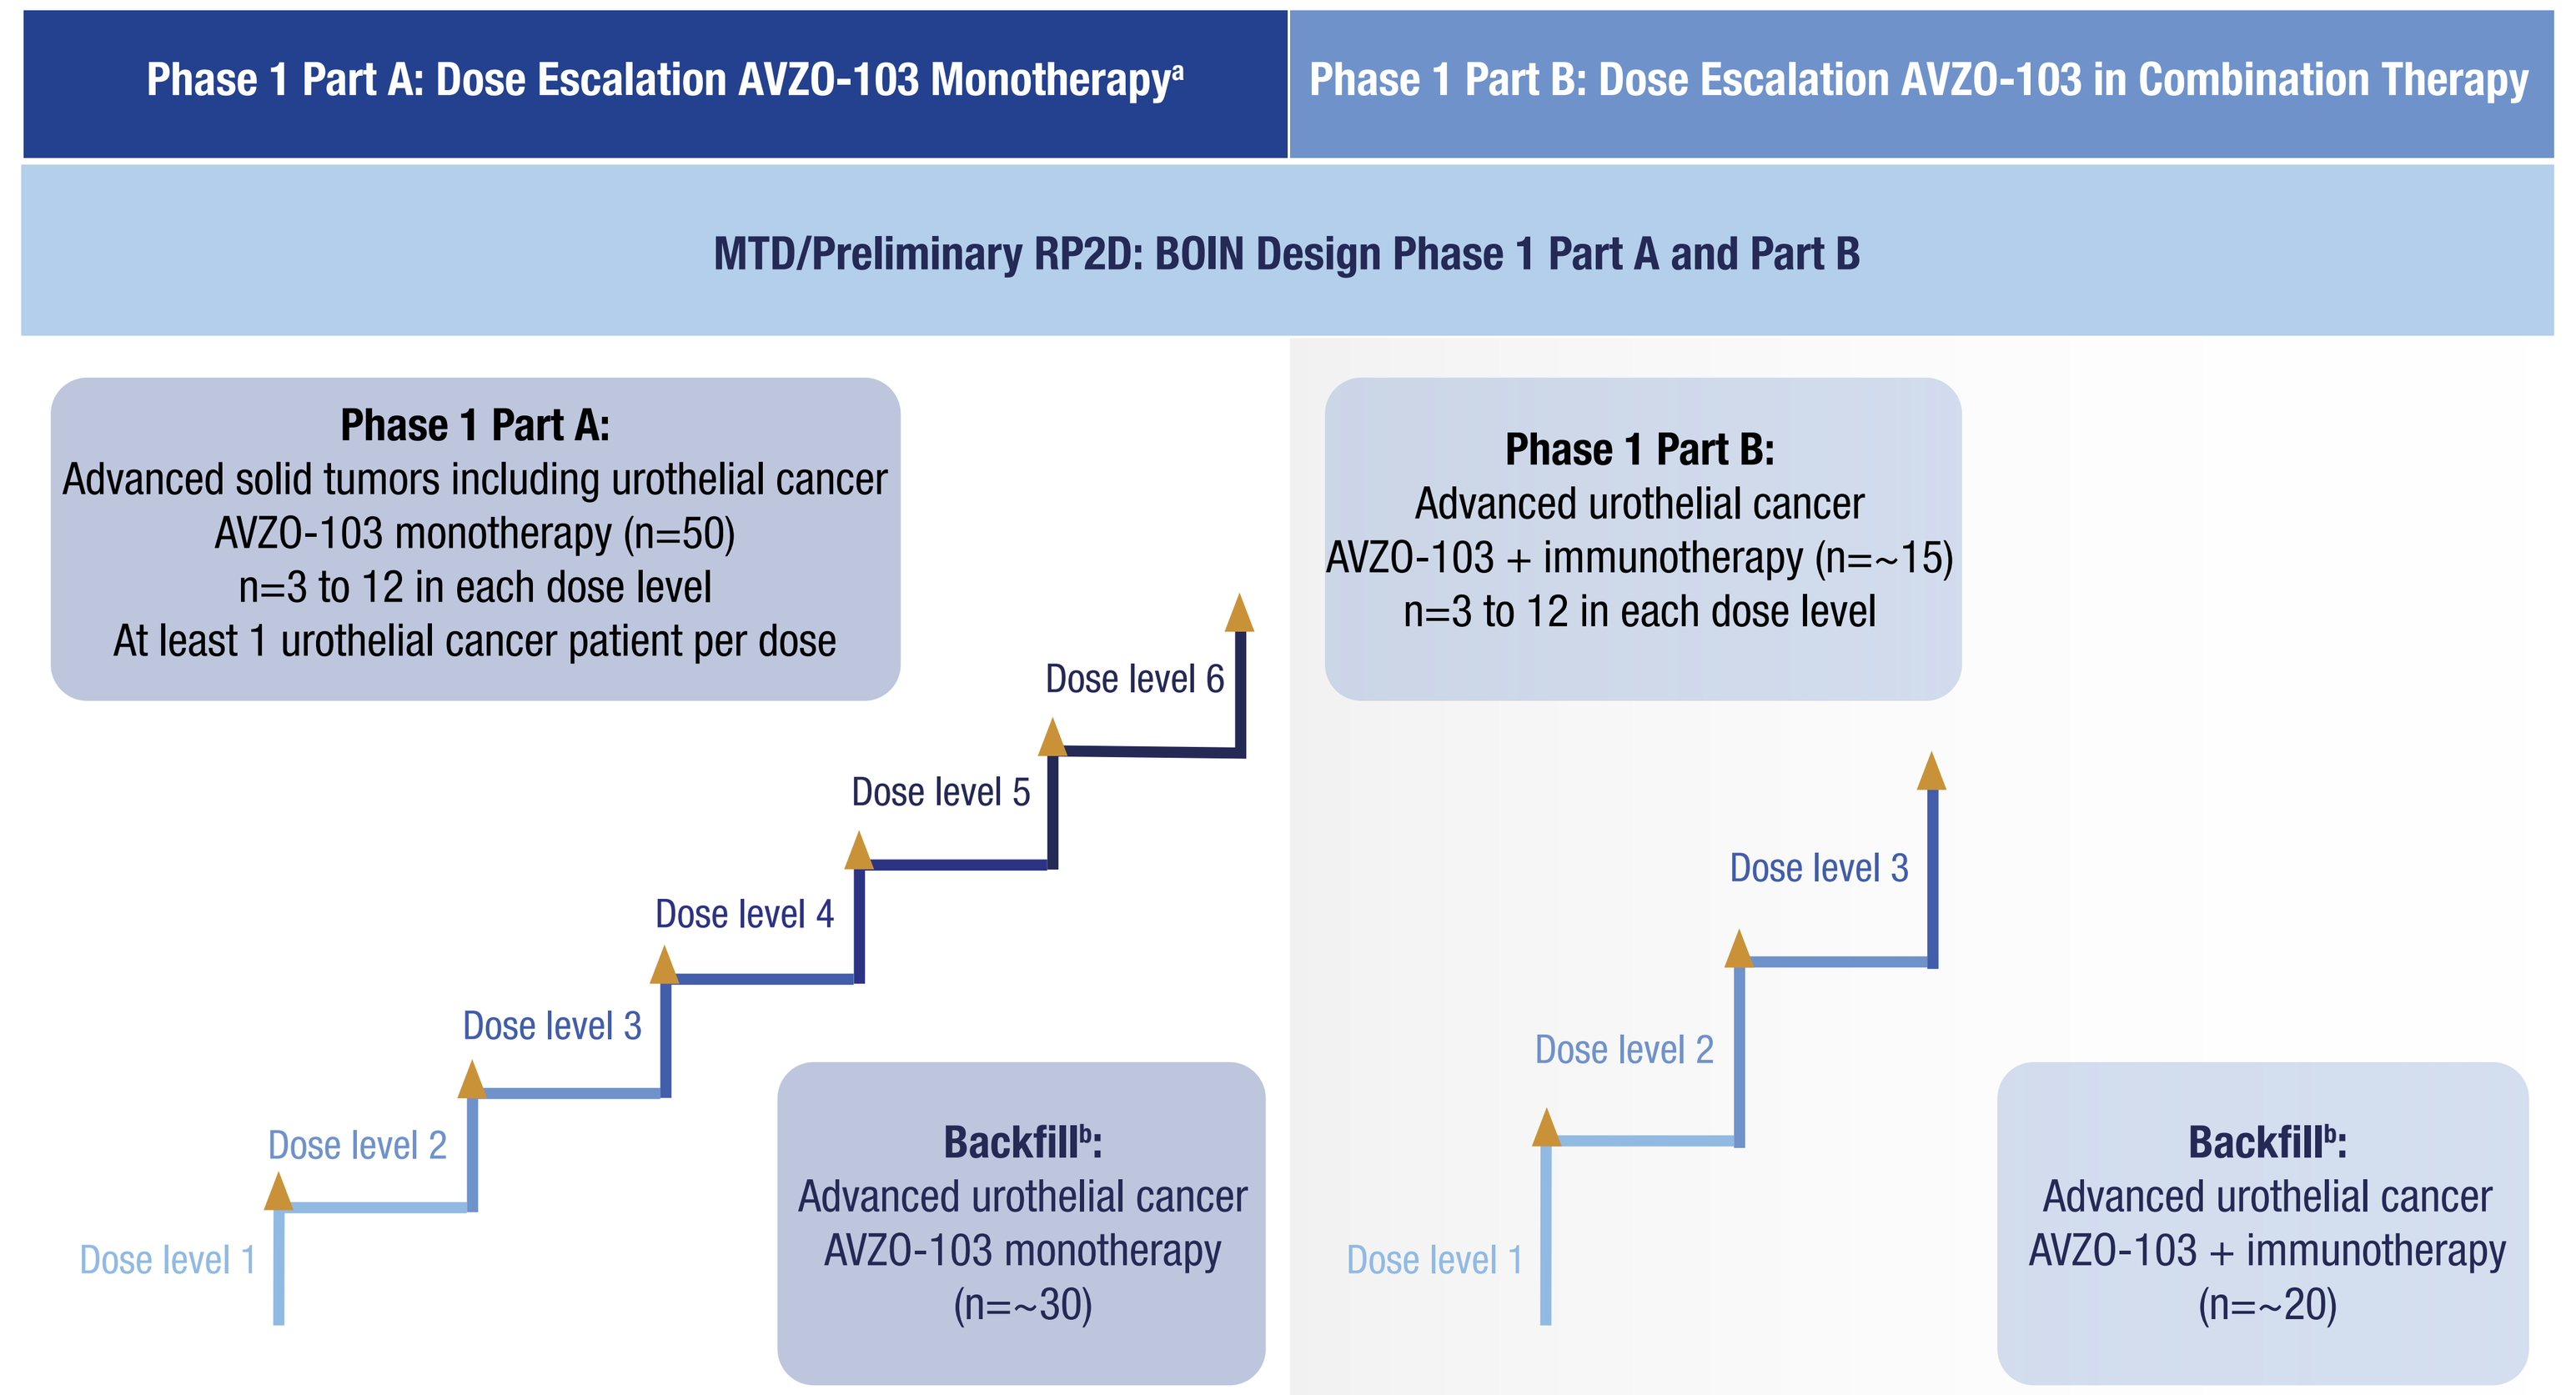
Phase 1 Part A: Dose Escalation AVZO-103 Monotherapy[a]
Phase 1 Part B: Dose Escalation AVZO-103 in Combination Therapy
MTD/Preliminary RP2D: BOIN Design Phase 1 Part A and Part B
Phase 1 Part A:
Advanced solid tumors including urothelial cancer
AVZO-103 monotherapy (n=50)
n=3 to 12 in each dose level
At least 1 urothelial cancer patient per dose
Dose level 1
Dose level 2
Dose level 3
Dose level 4
Dose level 5
Dose level 6
Backfill[b]:
Advanced urothelial cancer
AVZO-103 monotherapy
(n=~30)
Phase 1 Part B:
Advanced urothelial cancer
AVZO-103 + immunotherapy (n=~15)
n=3 to 12 in each dose level
Dose level 1
Dose level 2
Dose level 3
Backfill[b]:
Advanced urothelial cancer
AVZO-103 + immunotherapy
(n=~20)

[a]Advanced solid tumors that are known to express high Nectin4, including UC, cervical cancer, TNBC, SCCHN, NSCLC with EGFR mutations, and non-squamous NSCLC with no AGAs. [b]Backfill enrollment to further explore safety, PK, and pharmacodynamics, and efficacy in 1 or more dose levels that have been cleared by the SRC for tolerability.

AGA, actionable genomic alteration; **BOIN,** Bayesian Optimal Interval Design; **MTD,** maximum tolerated dose; **NSCLC,** non-small cell lung cancer; **PK,** pharmacokinetics; **RP2D,** recommended Phase 2 dose; **SCCHN,** squamous cell carcinoma of the head and neck; **SRC,** safety review committee; **TNBC,** Triple Negative Breast Cancer; **UC,** urothelial cancer.

Figure 2. Study design of AVZO-103-1001.[12]

KEY ELIGIBILITY CRITERIA[10,12]

Key Inclusion Criteria

- ≥18 years of age
- For Phase 1 Part A, histologically or cytologically confirmed locally advanced or metastatic select solid tumors[a] known to express Nectin4 for dose escalation and urothelial cancer in backfill, with the following prior therapy[b]:
 - ≤3 prior systemic lines of cytotoxic chemotherapy (≤2 for backfill patients) in the metastatic setting, including ADCs (non-topoisomerase inhibitor payloads)
 - Other therapies are permitted, including prior immunotherapy
- Measurable disease as assessed by investigator using Response Evaluation Criteria in Solid Tumors (RECIST) v1.1
- Eastern Cooperative Oncology Group (ECOG) Performance Status of ≤1 and a life expectancy of >3 months

Key Exclusion Criteria

- Known active brain metastases[c]
- Other active malignancy within 3 years prior to the first dose in this study[d]
- Participation in other studies involving investigational drugs within 2 weeks (or 4 weeks for biologicals) prior to the first dose of this study or completed 5 half-lives of the drug, whichever is shorter
- Received radiotherapy with a limited field of radiation within 7 days of the first dose of study treatment
- Unresolved Grade >1 toxicities from prior therapy as assessed by Common Terminology Criteria for Adverse Events v5.0, with exceptions of alopecia, vitiligo, and Grade ≤2 peripheral neuropathy
- Patients who previously received a prior ADC with a topoisomerase 1 inhibitor payload
 - Patients with TNBC are eligible if they have received ≤1 prior ADC with a topoisomerase 1 payload

[a]Includes UC, cervical cancer, TNBC, SCCHN, NSCLC with EGFR mutations, and non-squamous NSCLC with no actionable genomic alterations (confirmed by molecular report). [b]Patients with non-squamous NSCLC without a known actionable genomic alteration should have received platinum-based chemotherapy and anti–PD-(L)1 therapy (either concurrently or sequentially); patients with non-squamous EGFR-mutated NSCLC should have progressed on standard targeted therapies. [c]Patients with asymptomatic and treated brain metastases may participate if they are radiologically stable for at least 4 weeks prior to the first dose of this study and do not require steroid treatment. Patients with suspected or confirmed leptomeningeal disease are not eligible, even if treated. [d]Excludes adequately treated basal cell or squamous cell skin cancer, carcinoma in situ, colorectal adenomatous polyps, or other malignancies for which the patient is not on active anticancer therapy.

STUDY ENDPOINTS[10,12]

Primary Endpoints

- **MTD or preliminary RP2D** of AVZO-103 as monotherapy and in combination therapy
 - Based on the number of patients with a dose-limiting toxicity during the first cycle
- **Safety and tolerability of AVZO-103** as monotherapy and in combination therapy
 - Includes incidence and severity of treatment-emergent or serious adverse events
 - Monitoring changes in clinical laboratory values, including hematology and chemistry values, vital signs, electrocardiograms, dose interruptions, dose reductions, and dose intensity

Secondary Endpoints

- **Preliminary antitumor activity** of AVZO-103 using RECIST v1.1 as assessed by the Investigator
 - Objective response rate and duration of response
 - Disease control rate
 - Progression-free survival and overall survival
- **Pharmacokinetics of AVZO-103, total antibody, and free payload**
 - Parameters considered include maximum and minimum observed concentrations, time to maximum observed concentration, elimination half-life, area under the concentration-time curve (AUC) from time 0 to the last measurable concentration, AUC from time 0 to infinity, AUC from time 0 to the end of the dosing period, clearance, volume of distribution at steady state, and accumulation ratio
- **Immunogenicity** of AVZO-103
 - Based on incidence of anti-AVZO-103 antibodies

The study is active and will enroll patients at centers across North America, Europe, and the Asia-Pacific region.

The latest information on this study can be found at ClinicalTrials.gov, reference number NCT07193511.

REFERENCES

1. Moretto R, et al. *Br J Cancer*. 2023;128(7):1391-1399. doi: 10.1038/s41416-023-02180-7 **2.** Takano A, et al. *Cancer Res*. 2009;69(16):6694-6703. doi: 10.1158/0008-5472.CAN-09-0016 **3.** Derycke MS, et al. *Am J Clin Pathol*. 2010;134(5):835-845. doi: 10.1309/AJCPGXK0FR4MHIHB **4.** Stepan LP, et al. *J Histochem Cytochem*. 2011;59(7):701-710. doi: 10.1369/0022155411410430 **5.** PADCEV (enfortumab vedotin-ejfv). Prescribing Information. Astellas Pharma, Inc.;2025. Accessed January 8, 2026. https://astellas.us/docs/padcev_label.pdf **6.** Gogia P, et al. *Cancers (Basel)*. 2023;15(15):3886. doi: 10.3390/cancers15153886 **7.** Klümper N, et al. *Clin Cancer Res*. 2023;29(8):1496-1505. doi: 10.1158/1078-0432.CCR-22-1764 **8.** Nguyen TD, et al. *Cancers (Basel)*. 2023.15(3):713. doi: 10.3390/cancers15030713 **9.** Zhu Y, et al. *Cancer*. 2023;129(2):283-295. doi: 10.1002/cncr.34507 **10.** Avenzo Therapeutics. AVZO-103 Investigators' Brochure. 2025. **11.** Li W, et al. *Mol Cancer Ther*. 2025;24(2):163-175. doi: 10.1158/1535-7163.MCT-24-0588 **12.** Avenzo Therapeutics. AVZO-103-1001 Clinical Study Protocol. 2025.

ACKNOWLEDGMENTS

The authors thank participants and their caregivers, the investigators, and all the site staff who have participated and who are participating in this study. This study is sponsored by Avenzo Therapeutics. The authors would like to acknowledge Dr. Arlene O. Siefker-Radtke for her contributions to the publication. Medical writing assistance was provided by Bluprint Oncology and was funded by Avenzo Therapeutics.

CONTACT

Please contact presenting author Benjamin Garmezy at ben.garmezy@scri.com with questions or comments.